Airway Inflammation and clinical manifestations in subjects with mild asthma: time course of Human Rhinovirus (HRV) induced changes

John Efthimiou[1], Alex J Mann[2], Ganesh Balaratnam[2], Jane Gunter[3], Pawel Rucki[3], Chris Poll[2], Martin Johnson[2], Tony Lockett[3] [1]Respiratory Specialist Consultancy, Oxford, [2]hVIVO Services Ltd., [3]hVIVO Services Ltd. at the time of the study

RATIONALE:

The immune response to viral infections in patients with asthma has been shown to be different to that in healthy subjects. This clinical trial was performed to evaluate the optimum dose, safety and time course of clinical and associated inflammation changes of a nasally administered GMP manufactured HRV16 strain in subjects with mild atopic asthma.

METHODS:

hVIVO recruited 20 male and female subjects with mild atopic intermittent asthma, aged between 20 and 46 years, not taking inhaled corticosteroids, with low levels of serum neutralizing antibodies. Subjects had full health screens including full histories and physical examinations, pulmonary function and skin prick tests. Subjects were brought to quarantine a day before nasal inoculation and randomized to either a 10TCID50 dose of virus (n=13) or placebo (sham, diluent) (n=7).

Symptoms of upper respiratory tract (URT) (nasal discharge, nasal obstruction, sneezing, sore throat), lower respiratory tract (LRT) (cough, shortness of breath), and systemic illness (headache, malaise, muscle aches), were recorded three times a day throughout quarantine on self-reported subject diary cards that graded each symptom between 0 (none) and 3 (most severe). Asthma Control Questionnaires (ACQ) were completed at intervals throughout the study and used to evaluate the virus induced changes in asthma control subsequent to inoculation on Day 0. Peak Expiratory Flow (PEF) and expired nitric oxide levels (FeNO) were measured daily in the morning throughout quarantine.

Throughout the controlled quarantine period, regular timed samples were taken and assessments performed for 8 days, before discharge. Quantification of virus by Polymerase Chain Reaction (PCR) were performed on nasopharyngeal swabs. Blood samples were taken for serum inflammatory markers and blood cell counts regularly over the 8 days.

Following inoculation, subjects that did not become infected were excluded from analysis. One of the subjects in the placebo group was non-compliant with their asthma medication throughout quarantine and as such was removed from analysis. Data from the present study in subjects with asthma were also compared to data obtained from healthy subjects (given the same dose of virus) from a previous study conducted in a similar manner.

RESULTS:

Baseline characteristics

Baseline demographics and characteristics of the individuals are presented in Table 1.

Infection rate / virology, ACQ-7, PEF, and FeNO

Of the 13 subjects with asthma that were inoculated with virus, 11 (85%) were infected (confirmed by PCR, Figure 1a), 4 of the infected (36%) had clinically important rises (≥0.5) in their ACQ-7 score between Day 0 and Day 7 (Figure 2 & 3) and also had reductions in PEF from their baseline (9-19%).

Maximum viral titres were similar between subjects with mild asthma and healthy subjects (Figure 1a). However, at least half of the subjects with asthma had earlier rises in viral loads than healthy subjects.

While FeNO in the placebo inoculated asthma group declined over the quarantine, the level in the infected group remained at or above baseline (Figure 1d).

Upper and lower respiratory symptoms

The infected subjects with asthma had a more rapid onset of URT symptoms, than healthy subjects and had significantly higher daily URT symptoms when compared to asthma subjects given placebo over days 2, 3, and 4 (Figure 1b, all time points between Day 2 morning and Day 4 morning, p<0.01). In addition, maximal changes in URT symptoms were significantly higher in infected subjects with asthma when compared to asthma subjects given placebo (Figure 2a, p<0.05).

Infected subjects with asthma had elevated LRT symptoms from Day 3 onwards and had significantly higher LRT symptoms when compared to asthma subjects given placebo on the mornings of Day 3 and 5 (Figure 1c, p<0.05)

In addition, maximal changes in LRT symptoms were highest in infected subjects with asthma, followed by placebo subjects with asthma, and then healthy subjects (Figure 2b, infected subjects with asthma vs healthy infected subjects, p<0.05).

Biomarkers

Baseline blood eosinophil levels in infected asthma subjects were higher in those that had reduced asthma control (ACQ≥0.5 change) following infection (Figure 3, p<0.05).

Serum IP-10 levels (and a number of other viral infection related biomarkers) were upregulated in infected subjects with asthma compared to placebo on Days 2 to 5 (Figure 1e). In addition, maximal log2 fold rises in IP 10 between Day 0 and Day 8 were significantly higher in infected subjects with asthma compared to placebo (Figure 1f, p<0.005).

Statistics

Upper and lower respiratory symptoms comparisons between infected and placebo groups at each time point were performed using RM two-way ANOVA with Fishers LSD test.

Maximal upper and lower respiratory symptom comparisons between infected, placebo and healthy groups were performed using ANOVA.

Maximal IP-10 levels comparison of infected and placebo groups were performed using Mann Whitney non parametric two-tailed test comparing ranks.

ACQ and baseline blood eosinophil level comparisons of groups were performed using Mann Whitney non parametric two-tailed test comparing ranks.

Table 1: Baseline demographics & characteristics of subjects with asthma

Subject Number	Age	Sex	Baseline FEV1 % predicted	ACQ-7	Eosinophils 10^9 cells / mL	FeNO ppb	Serum IgE (U/mL)
Infected n=11							
1	21	F	107	0.29	0.29	10	129
3	24	M	87	0.57	0.12	11	206
5	27	F	84	0.57	0.24	45	19
6	32	M	72	0.57	0.39	63	32
7	30	M	88	0.86	0.09	27	7 $
8	21	M	96	0.43	0.54	63	128
10	23	M	74	0.57	0.52	55	30
12	26	M	76	0.43	0.28	41	82
15	22	M	97	0.57	0.30	95	133
16	23	M	99	0.57	0.21	43	15
19	35	M	79	1.00	0.15	75	72
Median / %	24	82% M	87	0.57	0.28	45	72
Placebo n=6							
4	25	M	106	0.00	0.24	24	3.05 $
9	20	M	104	0.14	0.45	20	1600
11	23	M	79	1.00	0.47	49	3.05 $
14	23	M	82	0.43	0.54	38	3.05 $
18	30	F	80	0.43	0.17	19	31
20	46	M	80	0.57	0.14	35	6.9
Median / %	24	83% M	82	0.43	0.35	35	6.9

$ Below LLOQ, value reported is ½ LLOQ

Figure 1: Changes in virology, symptoms and blood biomarkers after challenge: subjects with mild atopic asthma given either HRV (red) or placebo (blue), and healthy subjects given HRV (black).

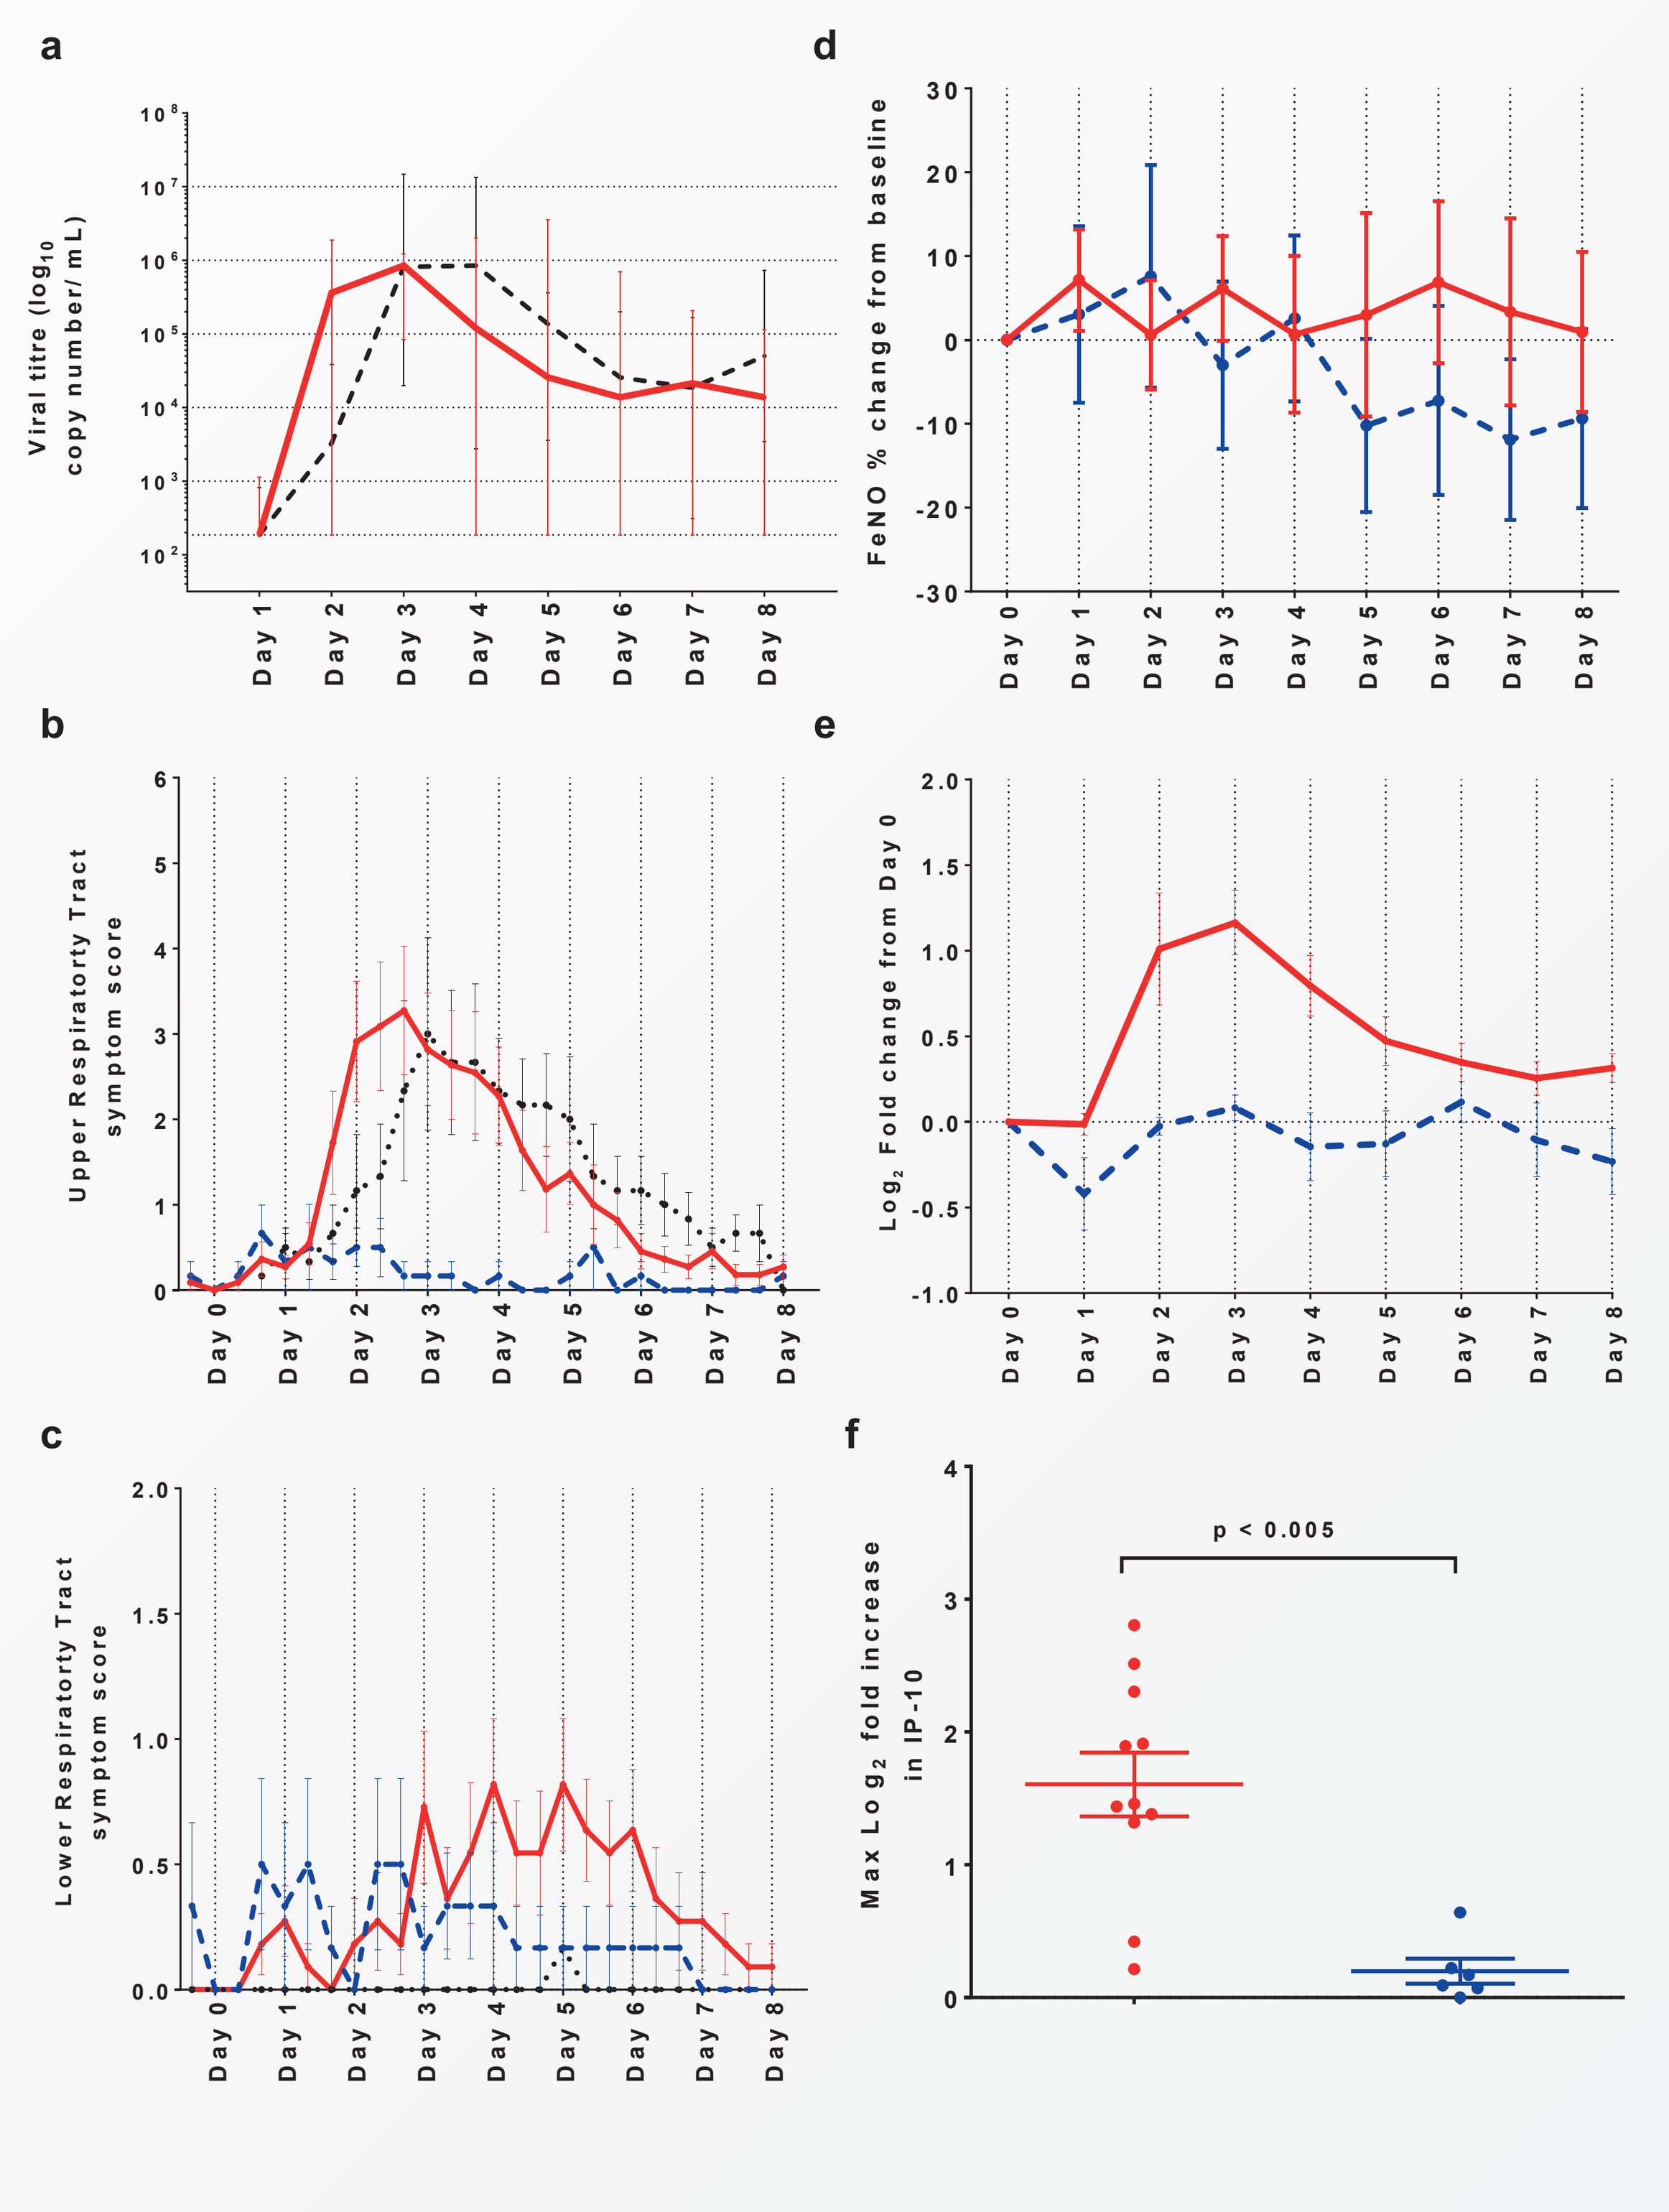

a) Median HRV viral loads in infected subjects with asthma (red, n=11) and healthy reference controls from a previous study given the same dose of virus (black, n=6). All values below the LLOQ were given the value of ½ LLOQ. Connecting lines represent the medians with interquartile range. b) Upper respiratory tract (URT) composite symptoms change from baseline Day 0 morning. c) Lower respiratory tract (LRT) composite symptoms change from baseline Day 0 morning. d) % change in expired nitric oxide levels (FeNO). e) IP-10 Log_2 fold change from baseline; f) Maximum Log_2 fold changes in IP-10 between Day 0 and Day 8. b) to e) connecting lines represent the means with standard error.

Figure 2: Maximum changes in symptom scores after HRV or diluent challenge: subjects with mild atopic asthma given HRV (red = controlled, green = reduced control) or virus diluent (blue), healthy subjects given HRV (black).

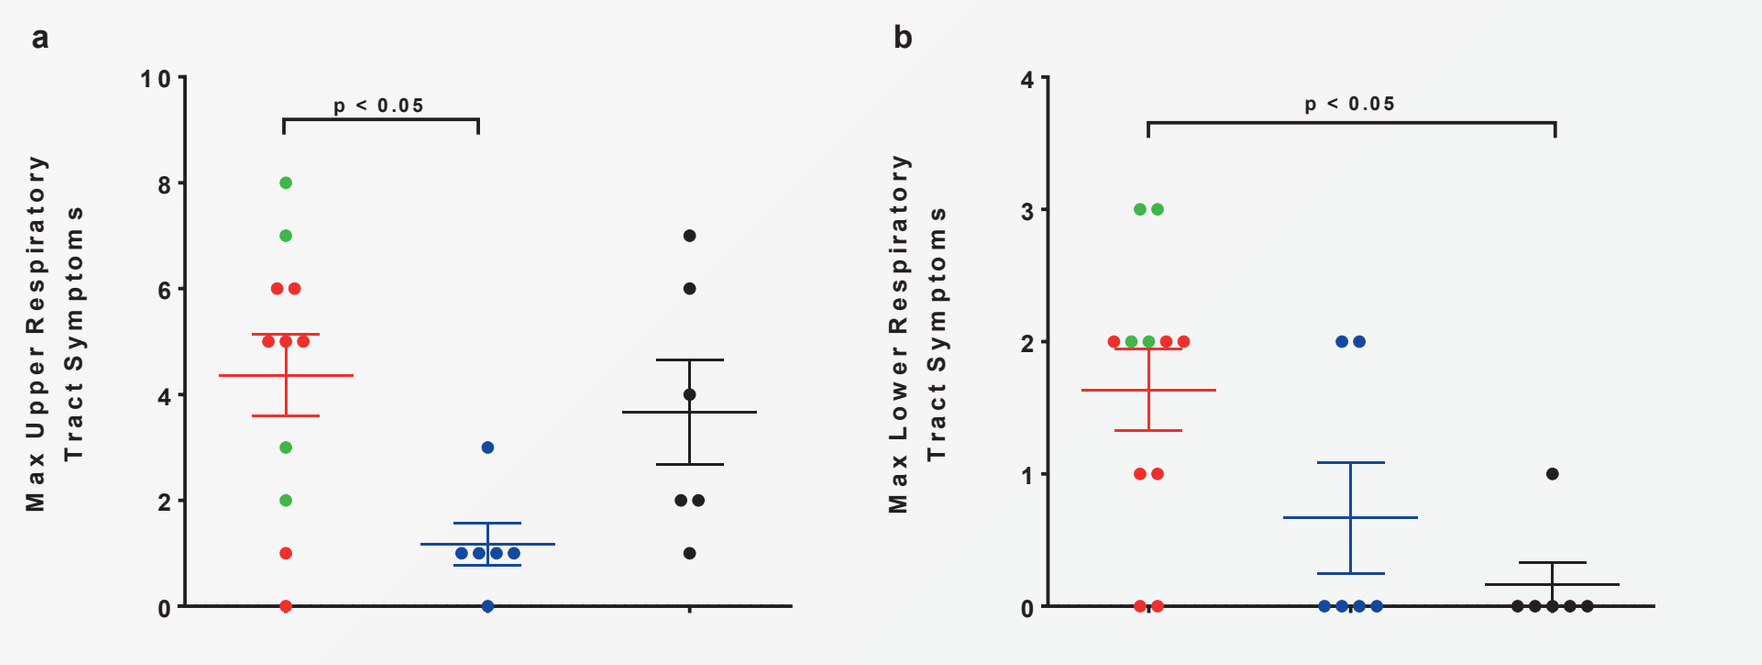

a) Maximal upper respiratory tract (URT) composite symptoms change from baseline; b) Maximal Lower respiratory tract (LRT) composite symptoms change from baseline. Asthma subjects infected (red, n=11) or given placebo (blue, n=6). Reference data from a previous study with the same dose of virus in healthy subjects (black). Asthma infected subjects that had a significant change in their asthma control (≥0.5 ACQ-7 rise from Day 0 to Day 7) are highlighted in green (n=4). All data presented are means and standard error.

Figure 3: Baseline (Day 0) blood eosinophils and reductions in control (increase of ≥0.5 ACQ) subsequent to HRV infection in mild atopic asthma. Subjects with mild atopic asthma given HRV (red = controlled, green = reduced control).

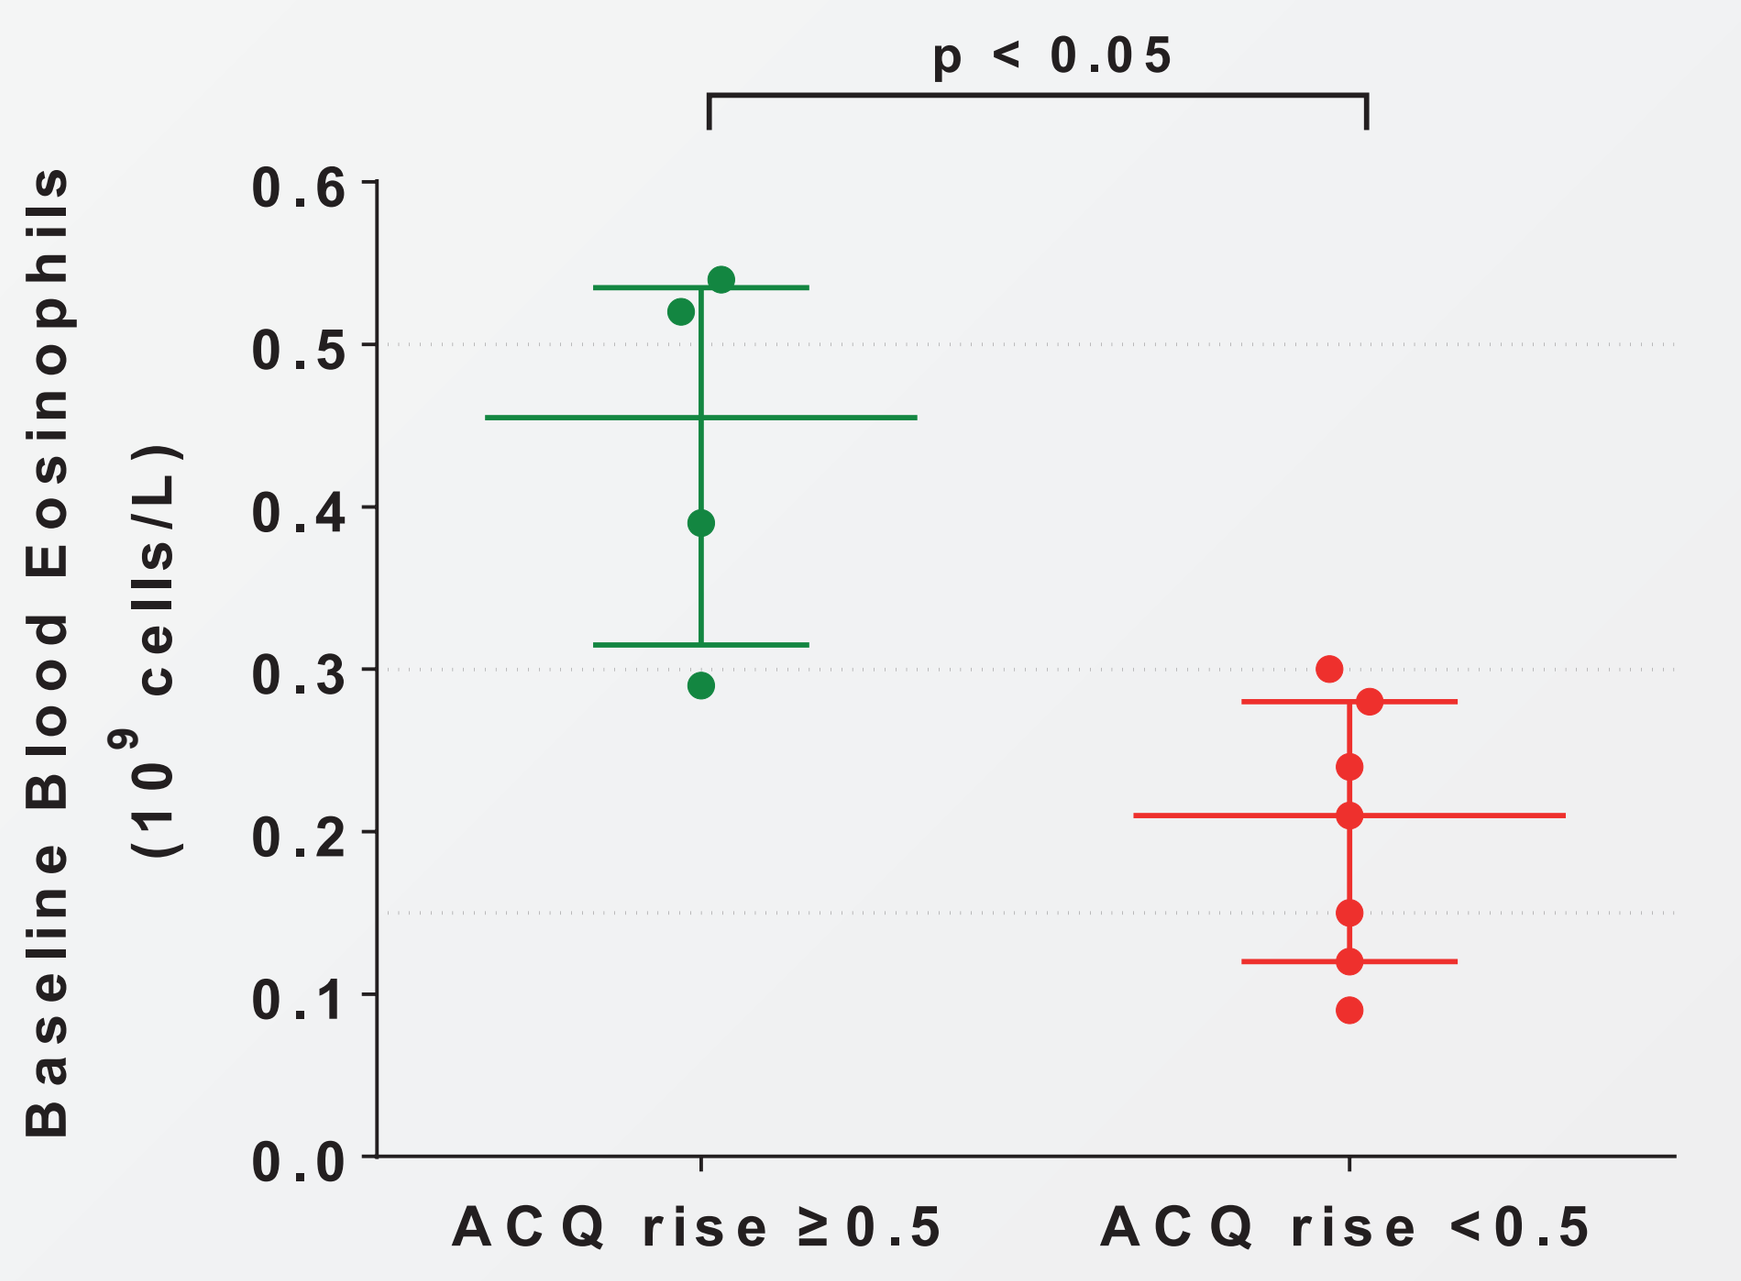
p < 0.05
Baseline Blood Eosinophils (10^9 cells/L)
ACQ rise ≥0.5
ACQ rise <0.5

Baseline (Day 0) peripheral blood eosinophil counts (10^9 cells/mL) in asthma infected subjects that had a significant change in their asthma control (≥0.5 ACQ-7 rise from Day 0 to Day 7) are highlighted in green (n=4), and those with less than 0.5 rise from baseline in red (n=7). Data presented are medians and interquartile range. For reference cut-points were included at 0.15, 0.3 and 0.5 counts (10^9 cells/mL).

CONCLUSION:

- Experimental HRV-16 infection in subjects with mild atopic asthma induced lower and upper respiratory tract symptoms, as well as rises in blood inflammatory markers (e.g. IP-10) compared to those inoculated with placebo.
- Of the 13 subjects with asthma that were inoculated with virus, 11 (85%) were infected (confirmed by PCR), 4 of the infected (36%) had clinically important increases in their ACQ-7 score (i.e. reduced control) with reductions in PEF compared to baseline.
- Infected asthma subjects that subsequently had reduced control as measured by ACQ, had higher baseline blood eosinophil counts than those that did not experience reduced control.
- A GMP HRV-16 strain of virus progressed safely from dose selection in healthy subjects to nasal inoculation and infection in subjects with mild atopic asthma.
- Robust, daily and comprehensive assessments and sampling using this experimental HRV-16 infection model in subjects with asthma within a quarantine setting, can aid in reducing data noise and confounders and allow for more precise and dynamic measurement of disease pathophysiology and drug efficacy.

www.hvivo.com | hVIVO Group PLC, Queen Mary BioEnterprises Innovation Centre, 42 New Rd, London E1 2AX. England. UK.

American Thoracic Society, International Conference
San Francisco, California. May 13 - 18 2016

Airway Inflammation and clinical manifestations in subjects with mild asthma: time course of Human Rhinovirus (HRV) induced changes

better treatments, faster

John Efthimiou[1], Alex J Mann[2], Ganesh Balaratnam[2], Jane Gunter[3], Pawel Rucki[3], Chris Poll[2], Martin Johnson[2], Tony Lockett[3] [1]Respiratory Specialist Consultancy, Oxford, [2]hVIVO Services Ltd., [3]hVIVO Services Ltd. at the time of the study

RATIONALE:

The immune response to viral infections in patients with asthma has been shown to be different to that in healthy subjects. This clinical trial was performed to evaluate the optimum dose, safety and time course of clinical and associated inflammation changes of a GMP manufactured HRV16 strain in subjects with mild atopic asthma.

METHODS:

hVIVO recruited 20 male and female subjects with mild atopic intermittent asthma, aged between 20 and 46 years, not taking inhaled corticosteroids, with low levels of serum neutralizing antibodies. Subjects had full health screens including full histories and physical examinations, pulmonary function and skin prick tests. Subjects were brought to quarantine a day before nasal inoculation and randomized to either a 10TCID50 dose of virus (n=13) or placebo (sham, diluent) (n=7).

Symptoms of upper respiratory tract (URT) (nasal discharge, nasal obstruction, sneezing, sore throat), lower respiratory tract (LRT) (cough, shortness of breath), and systemic illness (headache, malaise, muscle aches), were recorded three times a day throughout quarantine on self-reported subject diary cards that graded each symptom between 0 (none) and 3 (most severe). Asthma Control Questionnaires (ACQ) were completed at intervals throughout the study and used to evaluate the virus induced changes in asthma control subsequent to inoculation on Day 0. Peak Expiratory Flow (PEF) and expired nitric oxide levels (FeNO) were measured daily in the morning throughout quarantine.

Throughout the controlled quarantine period, regular timed samples were taken and assessments performed for 8 days, before discharge. Quantification of virus by Polymerase Chain Reaction (PCR) were performed on nasopharyngeal swabs. Blood samples were taken for serum inflammatory markers and blood cell counts regularly over the 8 days.

Following inoculation, subjects that did not become infected were excluded from analysis. One of the subjects in the placebo group was non-compliant with their asthma medication throughout quarantine and as such was removed from analysis. Data from the present study in subjects with asthma were also compared to data obtained from healthy subjects (given the same dose of virus) from a previous study conducted in a similar manner.

RESULTS:

Baseline characteristics

Baseline demographics and characteristics of the individuals are presented in Table 1.

Infection rate / virology, ACQ-7, PEF, and FeNO

Of the 13 subjects with asthma that were inoculated with virus, 11 (85%) were infected (confirmed by PCR, Figure 1a), 4 of the infected (36%) had clinically important rises (≥0.5) in their ACQ-7 score between Day 0 and Day 7 (Figure 2 & 3) and also had non-significant reductions in PEF from their baseline (9-19%).

Maximum viral titres were similar between subjects with mild asthma and healthy subjects (Figure 1a). However, at least half of the subjects with asthma had earlier rises in viral loads than healthy subjects.

While FeNO in the placebo inoculated asthma group declined over the quarantine, the level in the infected group remained above baseline (Figure 1d).

Upper and lower respiratory symptoms

The infected subjects with asthma had a more rapid onset of URT symptoms, than healthy subjects and had significantly higher daily URT symptoms when compared to asthma subjects given placebo over days 2, 3, and 4 (Figure 1b; all time points between Day 2 morning and Day 4 morning p<0.01). In addition, maximal changes in URT symptoms were significantly higher in infected subjects with asthma when compared to asthma subjects given placebo (Figure 2a, p<0.05).

Infected subjects with asthma had elevated LRT symptoms from Day 3 onwards and had significantly higher LRT symptoms when compared to asthma subjects given placebo on the mornings of Day 3 and 5 (Figure 1c; p<0.05).

In addition, maximal changes in LRT symptoms were highest in infected subjects with asthma, followed by placebo subjects with asthma, and then healthy subjects (Figure 2b, infected subjects with asthma vs healthy infected subjects p<0.05).

Biomarkers

Baseline blood eosinophil levels in infected asthma subjects were higher in those that had reduced asthma control (ACQ≥0.5 change) following infection (Figure 3; p<0.05).

Serum IP-10 levels (and a number of other viral infection related cytokines) were upregulated in infected subjects with asthma compared to placebo on Days 2 to 5 (Figure 1e). In addition, maximal log2 fold rises in IP 10 between Day 0 and Day 8 were significantly higher in infected subjects with asthma compared to placebo (Figure 1f; p<0.005).

Statistics

Upper and lower respiratory symptoms: comparisons between infected and placebo groups at each time point were performed using RM two-way ANOVA with Fishers LSD test.

Maximal upper and lower respiratory symptom comparisons between infected, placebo and healthy groups were performed using ANOVA.

Maximal IP-10 levels: comparison of infected and placebo groups were performed using Mann Whitney non parametric two-tailed test comparing ranks.

ACQ and baseline blood eosinophil level comparisons of groups were performed using Mann Whitney non parametric two-tailed test comparing ranks.

Table 1: Baseline demographics & characteristics of subjects with asthma

Subject Number	Age	Sex	Baseline FEV1 % predicted	ACQ-7	Eosinophils 109 cells / mL	FeNO ppb	Serum IgE (U/mL)
Infected n=11							
1	21	F	107	0.29	0.29	10	129
3	24	M	87	0.57	0.12	11	206
5	27	F	84	0.57	0.24	45	19
6	32	M	72	0.57	0.39	63	32
7	30	M	88	0.86	0.09	27	7$
8	21	M	96	0.43	0.54	63	128
10	23	M	74	0.57	0.52	55	30
12	26	M	76	0.43	0.28	41	82
15	22	M	97	0.57	0.30	95	133
16	23	M	99	0.57	0.21	43	15
19	35	M	79	1.00	0.15	75	72
Median / %	24	82% M	87	0.57	0.28	45	72
Placebo n=6							
4	25	M	106	0.00	0.24	24	3.05$
9	20	M	104	0.14	0.45	20	1600
11	23	M	79	1.00	0.47	49	3.05$
14	23	M	82	0.43	0.54	38	3.05$
18	30	F	80	0.43	0.17	19	31
20	46	M	80	0.57	0.14	35	6.9
Median / %	24	83% M	82	0.43	0.35	35	6.9

Figure 1: Changes in virology, symptoms and blood biomarkers after challenge: subjects with mild atopic asthma given either HRV (red) or placebo (blue), and healthy subjects given HRV (black).

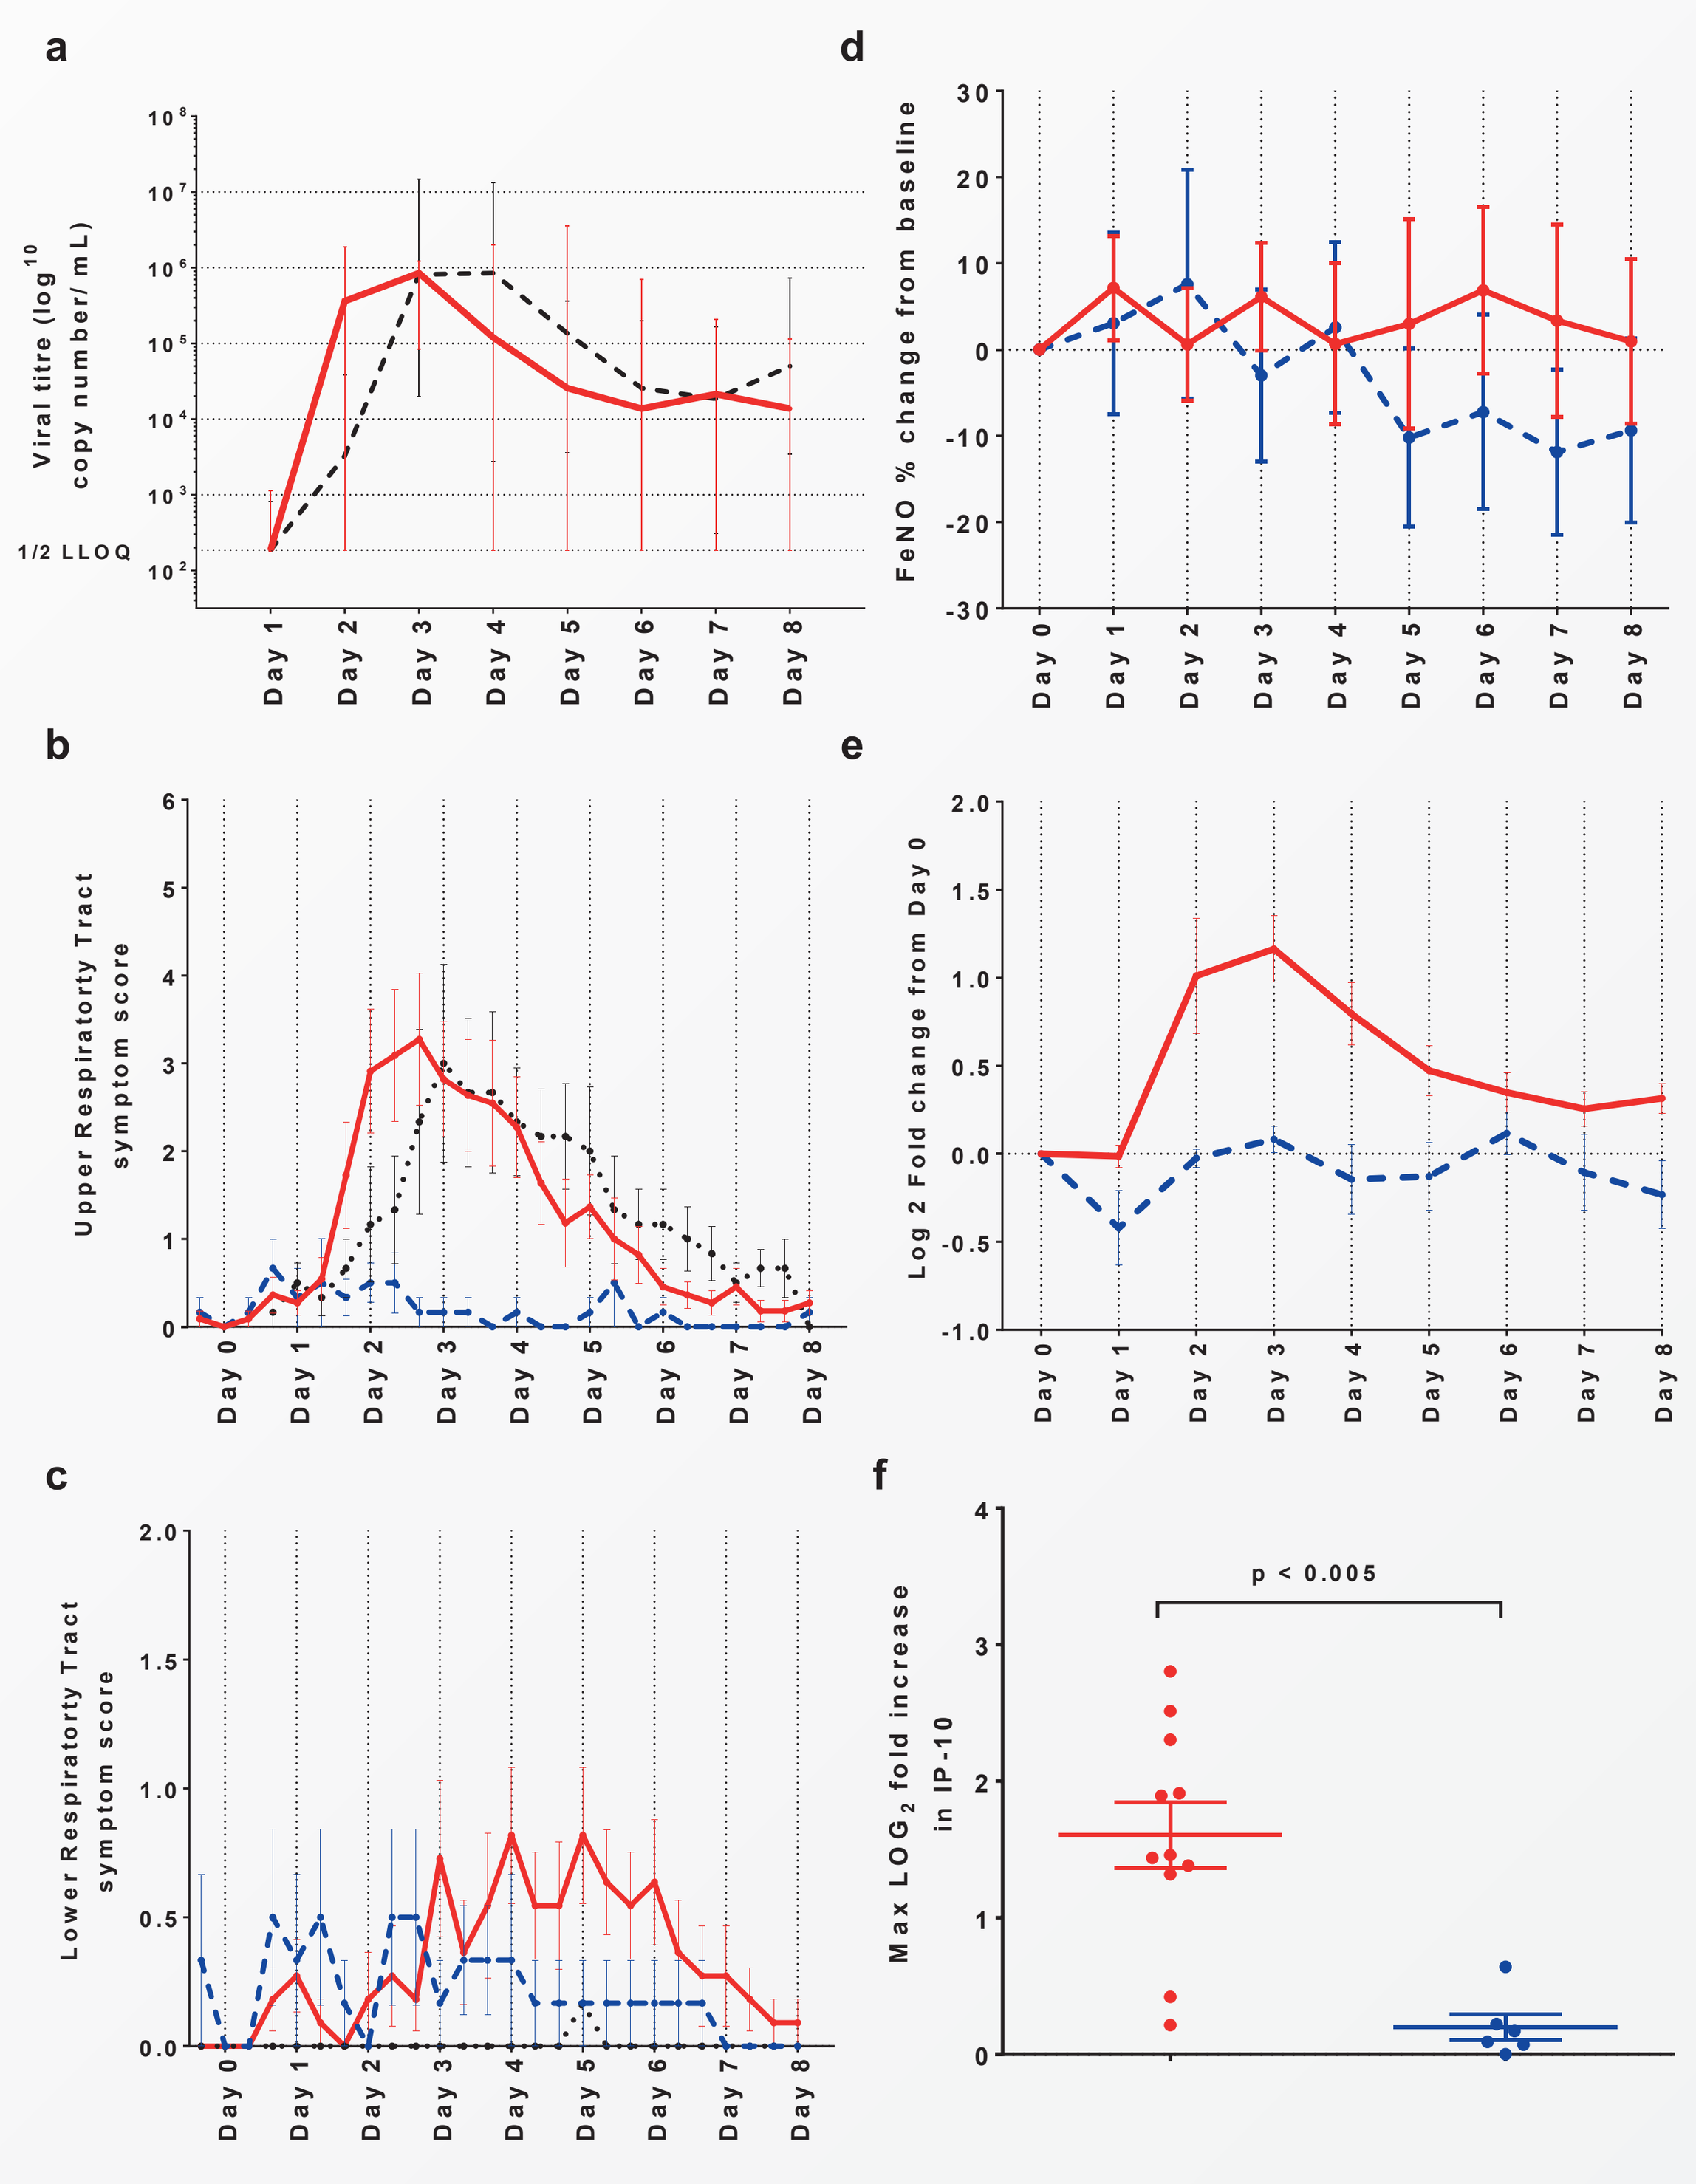
a
Viral titre (log10 copy number/ mL)
1/2 LLOQ
d
FeNO % change from baseline
b
Upper Respiratorty Tract symptom score
e
Log 2 Fold change from Day 0
c
Lower Respiratorty Tract symptom score
f
Max LOG2 fold increase in IP-10
p < 0.005

a) Median HRV viral loads in infected subjects with asthma (red, n=11) and healthy reference controls from a previous study given the same dose of virus (black, n=6). All values below the LLOQ were given the value of ½ LLOQ. Connecting lines represent the medians with interquartile range. b) Upper respiratory tract (URT) composite symptoms change from baseline Day 0 morning. c) Lower respiratory tract (LRT) composite symptoms change from baseline Day 0 morning. d) % change in expired nitric oxide levels (FeNO).e) IP-10 Log2 fold change from baseline; f) Maximum log2 fold changes in IP-10 between Day 0 and Day 8. b)-e) Connecting lines represent the means with standard error.

Figure 2: Maximum changes in symptom scores after HRV or diluent challenge: subjects with mild atopic asthma given HRV (red = controlled, green = reduced control) or virus diluent (blue), healthy subjects given HRV (black).

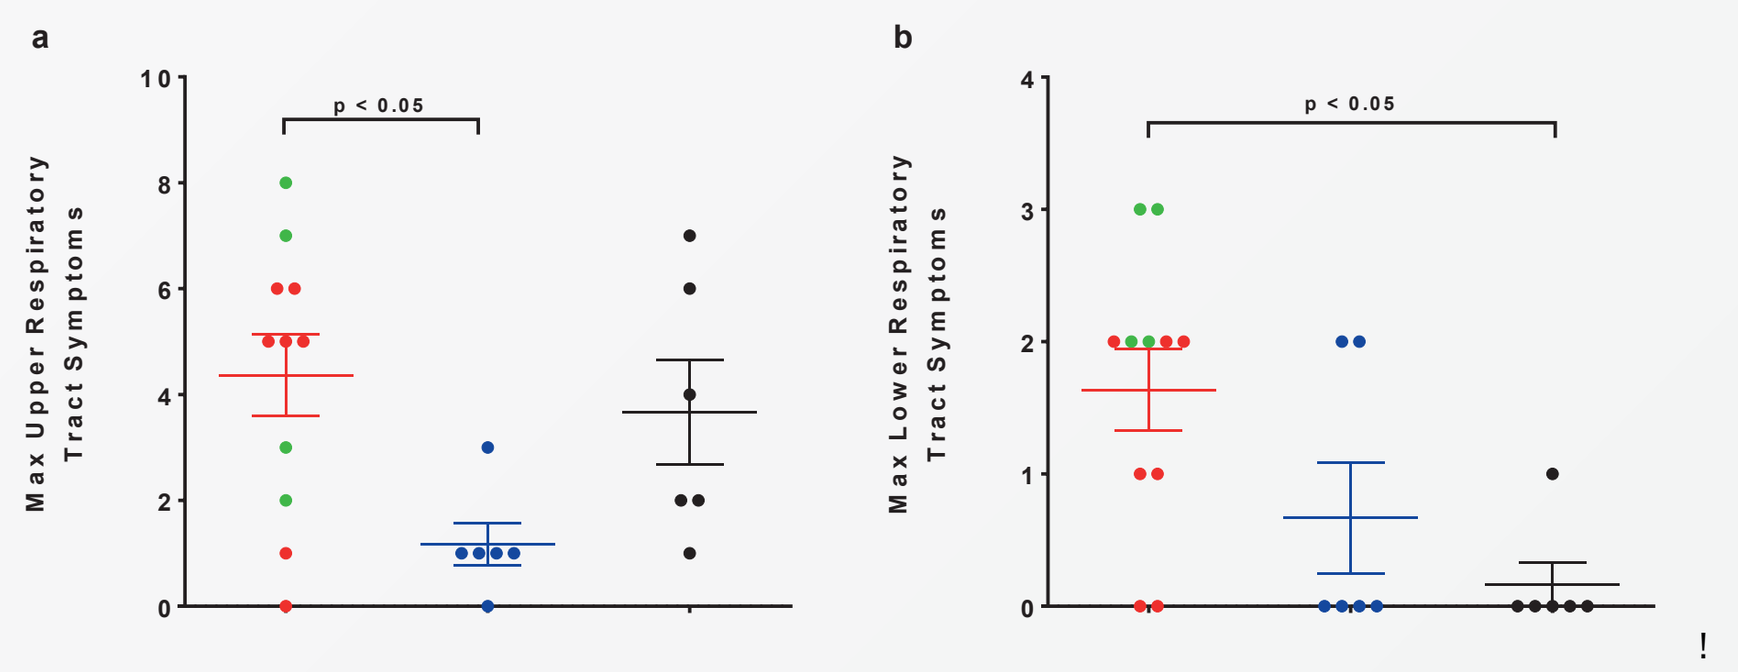
a
Max Upper Respiratory Tract Symptoms
p < 0.05
b
Max Lower Respiratory Tract Symptoms
p < 0.05

a) Maximal upper respiratory tract (URT) composite symptoms change from baseline; b) Maximal Lower respiratory tract (LRT) composite symptoms change from baseline. Asthma subjects infected (red, n=11) or given placebo (blue, n=6). Reference data from a previous study with the same dose of virus in healthy subjects (black). Asthma infected subjects that had a significant change in their asthma control (≥0.5 ACQ-7 rise from Day 0 to Day 7) are highlighted in green (n=4). All data presented are means and standard error.

Figure 3: Baseline (Day 0) blood eosinophils and reductions in control (increase of >0.5 ACQ) subsequent to HRV infection in mild atopic asthma. Subjects with mild atopic asthma given HRV (red = controlled, green = reduced control).

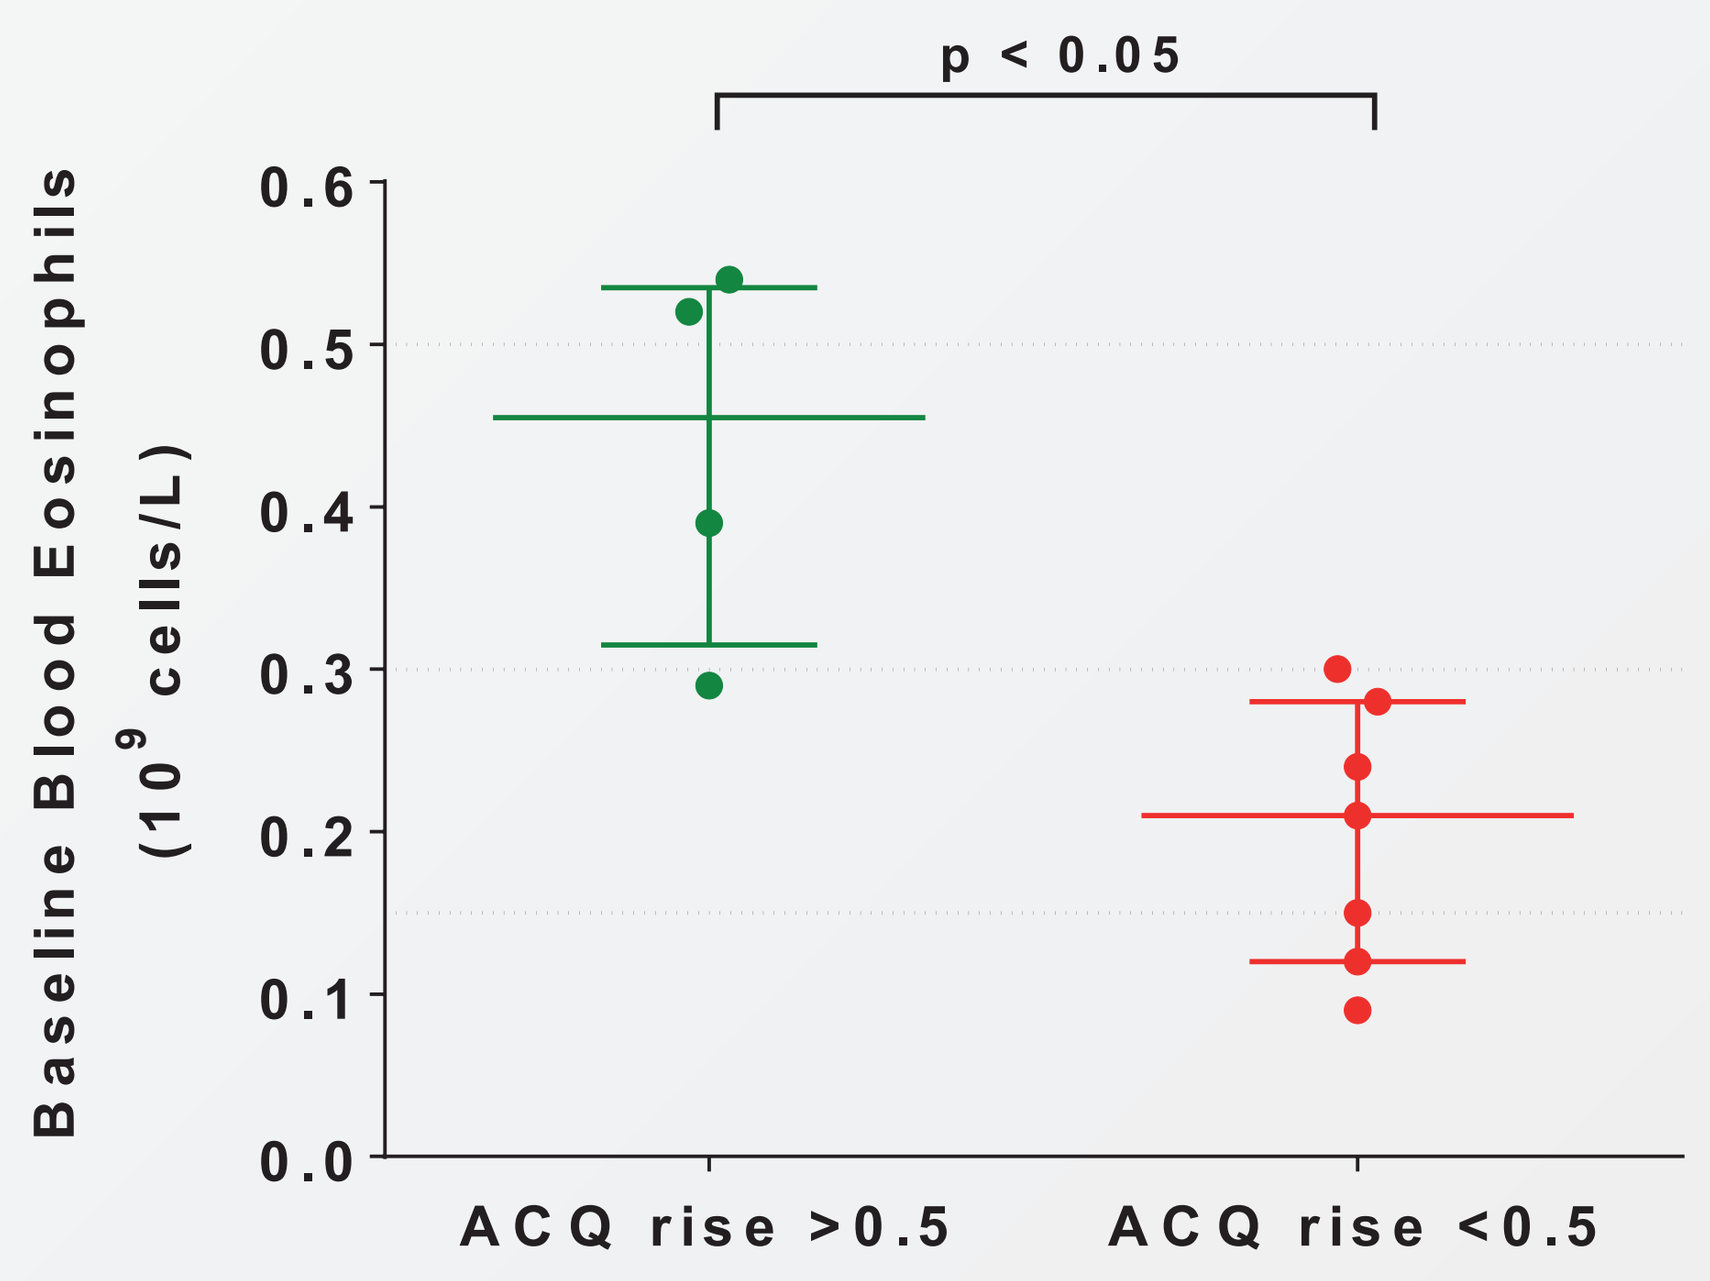
p < 0.05
Baseline Blood Eosinophils (10^9 cells/L)
ACQ rise >0.5
ACQ rise <0.5

Baseline (Day 0) peripheral blood eosinophil counts (109 cells/mL) in asthma infected subjects that had a significant change in their asthma control (≥0.5 ACQ-7 rise from Day 0 to Day 7) are highlighted in green (n=4), and those with less than 0.5 rise from baseline in red (n=7). Data presented are medians and interquartile range. For reference cut-points were included at 0.15, 0.3 and 0.5 counts (109 cells/mL).

CONCLUSION:

- Experimental HRV-16 infection in subjects with mild atopic asthma induced lower and upper respiratory tract symptoms, as well as rises in blood inflammatory markers (eg IP-10) compared to those inoculated with placebo.
- Of the 13 subjects with asthma that were inoculated with virus, 11 (85%) were infected (confirmed by PCR), 4 of the infected (36%) had clinically important increases in their ACQ-7 score (i.e. reduced control) with reductions in PEF compared to baseline.
- Infected asthma subjects that subsequently had reduced control as measured by ACQ, had higher baseline blood eosinophil counts than those that did not experience reduced control.
- A GMP HRV-16 strain of virus progressed safely from dose selection in healthy subjects to nasal inoculation and infection in subjects with mild atopic asthma.
- Robust, daily and comprehensive assessments and sampling using this experimental HRV-16 infection model in subjects with asthma within a quarantine setting, can aid in reducing data noise and confounders and allow for more precise and dynamic measurement of disease pathophysiology and drug efficacy.

www.hvivo.com | hVIVO Group PLC, Queen Mary BioEnterprises Innovation Centre, 42 New Rd, London E1 2AX. England. UK.

American Thoracic Society, International Conference
San Francisco, California. May 13 - 18 2016